THE SALINITY EFFECT ON MORPHOLOGY AND PIGMENTS CONTENT IN THREE PAULOWNIA CLONES GROWN *EX VITRO*

Kamelia Miladinova*, Katya Ivanova, Teodora Georgieva*, Mariya Geneva***, Yuliana Markovska****

** BioTree Ltd, 8 Iliensko shosse str., 1220 Sofia, Bulgariaя;*
*** Faculty of Biology, University of Sofia, 8 Dragan Tsankov Blvd., Sofia 1164, Bulgaria;*
**** Institute of Plant Physiology and Genetics, Bulgarian Academy of Sciences, Acad. Georgy Bonchev str., bl. 21, Sofia 1113, Bulgaria;*

ABSTRACT

We evaluated the effect of salt stress on growth parameters, chlorophyll a, chlorophyll b and carotenoids content in *Paulownia* clones (*P. elongata x fortunei x elongata* - T2, *P. elongata x elongata* - T4, *P. elongata x kawakarnii* - EK) grew as hydroponic culture at three levels of salinity, 50 mmol.l^{-1}, 100 mmol.l^{-1}, 200 mmol.l^{-1} sodium chloride (NaCl) solution. The root and stem length, as well as leaf number and total leaf area of T2 clone were reduced insignificantly in comparison with these of T4 and EK clones during NaCl treatment. Control of T2 clone was characterized with approximately twice higher total dry mass per plant than EK and T4 clones. The root dry mass to shoot dry mass ratios of three clones changed in a different manner with increasing salinity levels and was highest in EK clone at 200 mmol.l^{-1} NaCl, followed by T4 and T2. The total leaf area showed the capability of a plant in forming of photosynthetic surface and was reduced more in T4 and EK clones under salt stress. The chlorophyll a to chlorophyll b ratio as well as the green to yellow pigments ratio increased only in the leaves of T4 clone. Our results suggest that T2 clone was more tolerant to salt stress than EK and T4 clones.

MATERIAL AND METHODS

Plant material. Seeds and *in vivo* explants from the species *P. elongata* and their hybrids with *P. fortunei* and *P. kawakarnii* were used for developing of *in vitro* multiplication protocol. For induction of shoots, explants were cultured on Murashige and Skoog (MS) nutrient medium included 2.5% (w/v) sucrose, 0.8% (w/v) agar and vitamins. For shoots multiplication MS medium supplemented with 4.439 μM 6-benzylaminopurine (BAP) and 0.537 μM indolilacetic acid (IAA) was used. After multiplication the shoots were transferred to rooting medium based on half strength basal salts MS medium, 2 % sucrose, 6 % agar and vitamins supplemented with 4.92 μM indole-3-butyric acid (IBA) and 1.075 μM IAA. The pH of all media was adjusted to 5.7 using 0.1 N HCl and 0.1 N NaOH before autoclaving. All cultures were incubated under controlled conditions - 16 h photoperiod, light intensity of 35 μmol m^{-2} s^{-1} and 24/18±1°C day/night temperature. After three weeks of rooting, the shoots were rinsed with 1.5 ml/l Proplant solution.

Hydroponic experiment. The experiments were set as four treatments including control, each treatment with 3 replications. The uniform explants were selected and transplanted to polyethylene vessels containing 1.2 l of 1/4 Hellriegel solution (Hellriegel, 1898) with an addition of A-Z microelements after Hoagland (pH 5.9) in growth chamber with a 16-h photoperiod (PAR 100 μmol m^{-2} s^{-1} on the upper leaf surface, 25/23±1 °C day/night temperature, relative humidity 60/70%). Each vessel contained two explants which represented one replication. After 21 days of cultivation the plants were transferred to 1/2 Hellriegel solution with the addition of A-Z microelements (pH 5.9). The salt treatment was applied on the 48th day after transplanting of explants when the plants had adapted to the conditions of 1/2 Hellriegel nutrient solution and 0 (control), 50, 100, and 200 mmol.l^{-1} NaCl was added. The solutions were aerated every day and were changed every 3 d to prevent depletion of nutrients and NaCl. Plants were harvested after 10 d of treatment. Toxicity symptoms (e.g. discoloration, pigmentation, yellowing and stunting) were assessed by eye through-out the experiment.

Measurement of plant growth. At the end of the experiment the plant samples were collected, washed with tap water and rinsed with distilled water before being separated into leaf, petiole, stem and root and fresh mass of each plant sample were measured gravimetrically. Dry mass of shoots and roots were determined after oven-drying (60^oC) for 2 days until constant weight was obtained. Leaf area was calculated using software program SigmaScan Pro 5.

Pigment determination. Samples were collected from the fully developed leaves of three plants grown in varying concentrations of NaCl. Samples of the control plants of three *Paulownia* clones were also collected at these times. For pigment extraction, 1-3 mg of freeze - dried material from the middle part of fully developed leaves of each plant was extracted with 5 ml 80% acetone and filtered through a glass filter G4. The pigment content was determined spectrophotometrically (Lichtenthaler,1987).

Statistical analysis. All values reported in this work were mean of at least three independent experiments. The mean values ±SD and exact number of experiments are given in the tables. The significance of differences between control and each treatment was analyzed by Fisher LSD test (P≤0.05) after performing ANOVA multifactor analysis.

Conclusion

We examined the physiological indicators in order to develop practicable strategie for selecting salt tolerant clones of *Paulownia* (*P. elongata x fortunei x elongata* - T2, *P. elongata x elongata* - T4, *P. elongata x kawakarnii* - EK), which were produced by BioTree Ltd., Bulgaria. Control of T2 clone was characterized with approximately twice higher total dry mass per plant than EK and T4 clones.The root and stem length, as well as leaf number and total leaf area of T2 clone were reduced insignificantly in comparison with these of T4 and EK clones during treatment with increasing concentrations of NaCl from 50 mmol.l^{-1} to 200 mmol.l^{-1}. The chlorophyll and carotenoid contents in the leaves of three clones remain relatively invariable, but chlorophyll a to chlorophyll b ratio as well as the green to yellow pigments ratio increased only in the leaves of T4 clone. Probably investigated three clones possessed different ways of adaptation of the photosynthetic apparatus to the action of salt. Our results suggest that T2 clone was more tolerant to salt stress than EK and T4 clones.

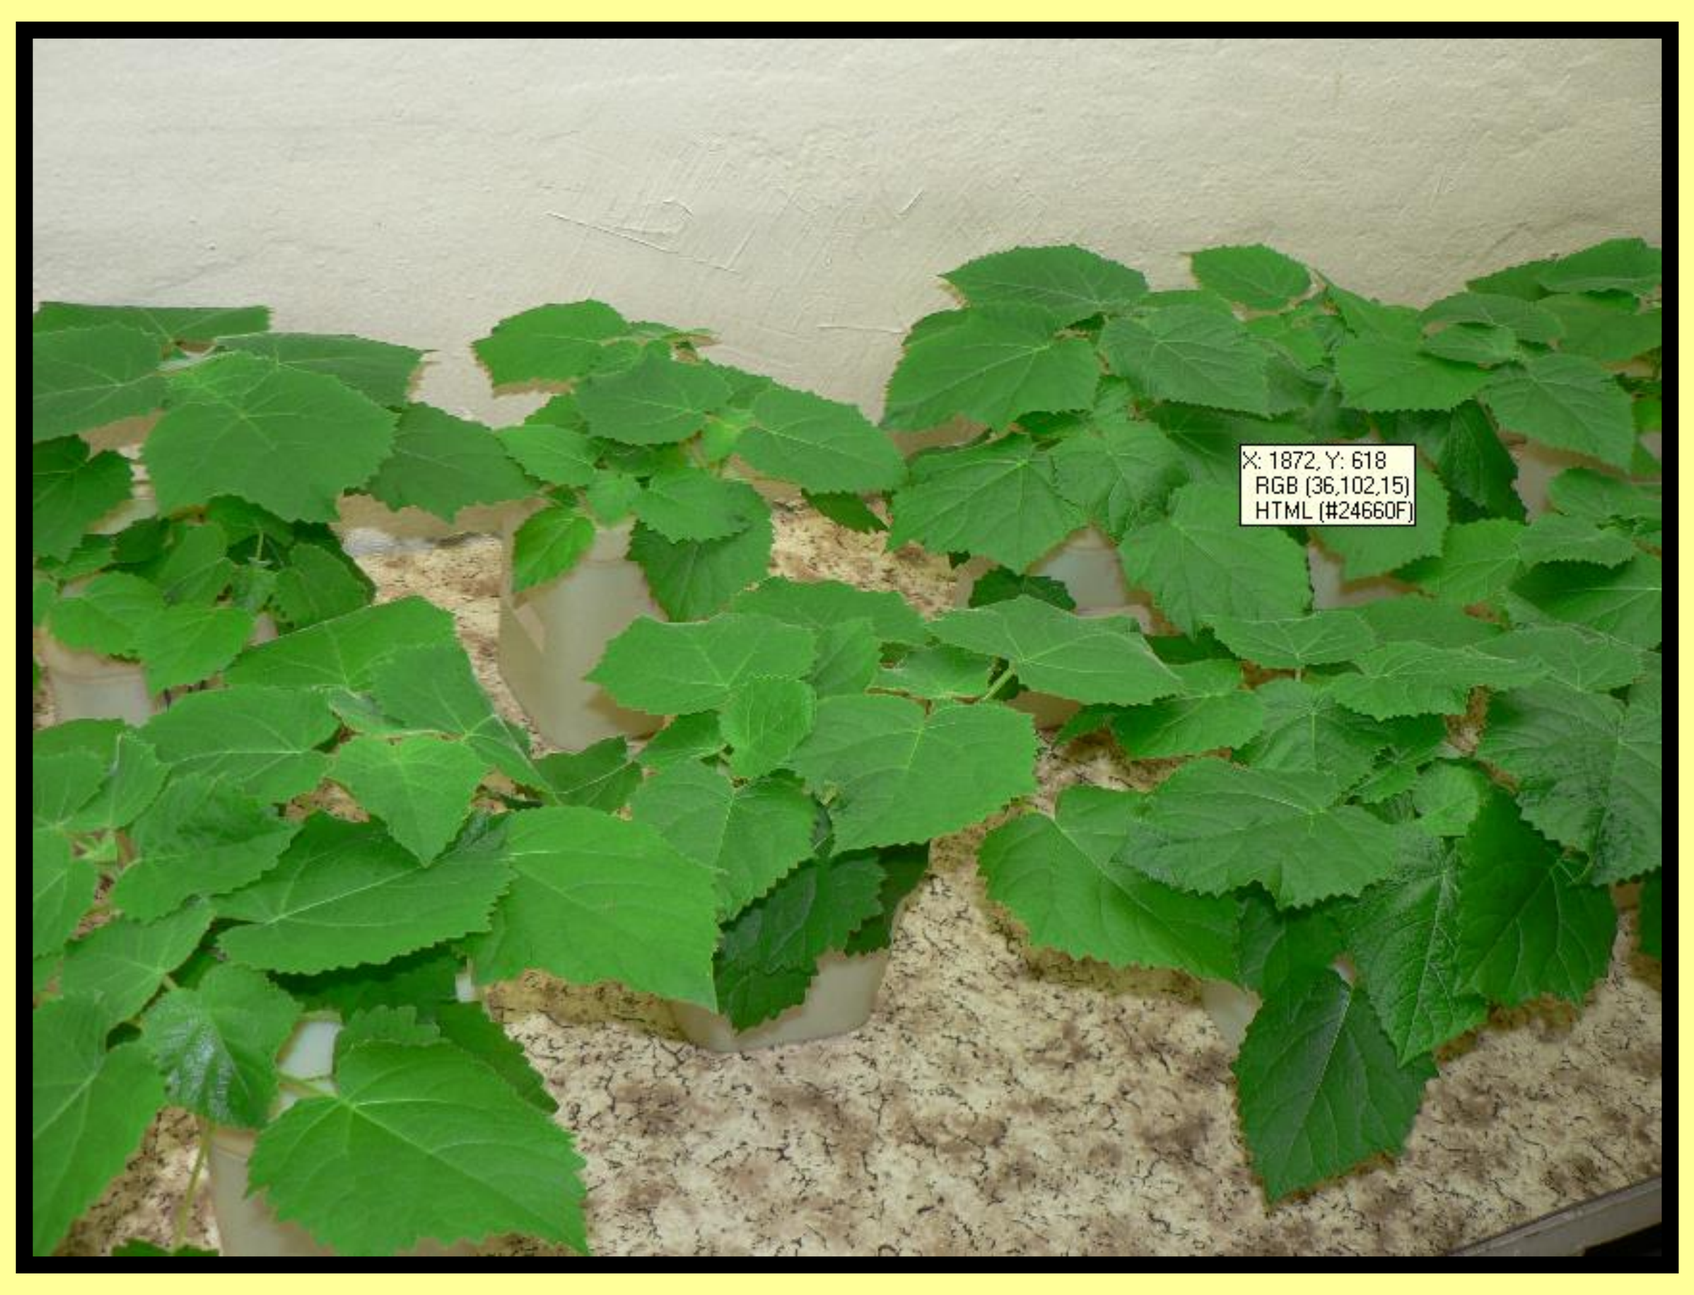

Treatments	Root length [cm]	Stem length [cm]	Leaf number	Total leaf area [cm^2 plant^{-1}]	Total DW [g plant^{-1}]	root DW/ shoot DW
T4						
Control	20.0 ± 3.0^{a}	8.0 ± 0.4^{a}	12.0 ± 2.9^{a}	411.7 ± 11.8^{a}	0.514 ± 0.038^{a}	0.168
50 mM.l^{-1} NaCl	16.1 ± 3.8^{b}	6.6 ± 1.8^{b}	12.0 ± 2.5^{a}	294.8 ± 28.4^{b}	0.658 ± 0.078^{b}	0.251
100 mM l^{-1} NaCl	24.5 ± 4.9^{c}	7.3 ± 0.2^{c}	10.0 ± 0.7^{b}	379.1 ± 46.1^{c}	0.736 ± 0.026^{c}	0.224
200 mM l^{-1} NaCl	17.8 ± 3.9^{d}	5.6 ± 2.0^{d}	9.0 ± 1.0^{c}	188.6 ± 19.8^{d}	0.324 ± 0.058^{d}	0.269
T2						
Control	19.3 ± 5.5^{a}	8.8 ± 1.2^{a}	12.0 ± 1.0^{a}	400.0 ± 61.4^{a}	1.034 ± 0.041^{a}	0.163
50 mM l^{-1} NaCl	17.5 ± 3.8^{b}	8.5 ± 1.3^{a}	12.0 ± 0.0^{a}	415.9 ± 80.8^{a}	0.826 ± 0.036^{b}	0.175
100 mM l^{-1} NaCl	15.3 ± 3.9^{c}	7.6 ± 0.9^{b}	12.0 ± 2.5^{a}	329.6 ± 12.1^{b}	0.608 ± 0.024^{c}	0.191
200 mM l^{-1} NaCl	16.2 ± 4.1^{d}	9.6 ± 1.7^{c}	8.0 ± 2.5^{b}	253.4 ± 61.5^{c}	0.458 ± 0.028^{d}	0.168
EK						
Control	17.8 ± 3.0^{a}	7.9 ± 2.3^{a}	8.0 ± 1.7^{a}	338.0 ± 68.8^{a}	0.616 ± 0.050^{a}	0.221
50 mM l^{-1} NaCl	14.4 ± 1.9^{b}	7.9 ± 2.3^{a}	11.0 ± 2.2^{b}	327.2 ± 32.3^{a}	0.683 ± 0.071^{b}	0.233
100 mM l^{-1} NaCl	10.7 ± 2.1^{c}	6.6 ± 2.5^{b}	9.0 ± 2.1^{c}	249.9 ± 27.9^{b}	0.454 ± 0.053^{c}	0.231
200 mM l^{-1} NaCl	13.6 ± 1.5^{d}	6.5 ± 1.1^{c}	9.0 ± 0.0^{c}	198.6 ± 10.7^{c}	0.341 ± 0.020^{d}	0.270

Treatments	Chl a / chl b	Green / yellow
T4		
Control	1.261 ± 0.31^{a}	13.029 ± 1.31^{a}
50 mM l^{-1} NaCl	1.456 ± 0.12^{b}	11.282 ± 1.24^{b}
100 mM l^{-1} NaCl	1.456 ± 0.42^{b}	12.556 ± 1.24^{c}
200 mM l^{-1} NaCl	1.656 ± 0.42^{c}	8.212 ± 0.73^{d}
T2		
Control	1.638 ± 0.42^{a}	8.540 ± 0.78^{a}
50 mM l^{-1} NaCl	1.573 ± 0.42^{b}	7.698 ± 0.60^{b}
100 mM l^{-1} NaCl	1.899 ± 0.21^{c}	9.903 ± 0.86^{c}
200 mM l^{-1} NaCl	1.638 ± 0.22^{d}	8.975 ± 0.75^{d}
EK		
Control	2.069 ± 0.22^{a}	4.246 ± 0.38^{a}
50 mM l^{-1} NaCl	1.856 ± 0.21^{b}	5.074 ± 0.47^{b}
100 mM l^{-1} NaCl	1.725 ± 0.15^{c}	5.774 ± 0.49^{c}
200 mM l^{-1} NaCl	1.819 ± 0.34^{d}	6.234 ± 0.54^{d}

Table 1. Mean values ± SD (n = 5-6) of the root and stem length, leaf number, total leaf area, total dry weight and root/shoot ratios of *Paulownia* clones (T4, T2, EK), grown as hydroponic culture in response to salt stress

Table 2. Mean values ± SD (n = 5-6) chlorophyll a/chlorophyll b and green/ yellow pigment ratios in the leaves of Paulownia clones (T4, T2, EK), grown as hydroponic culture in response to salt stress

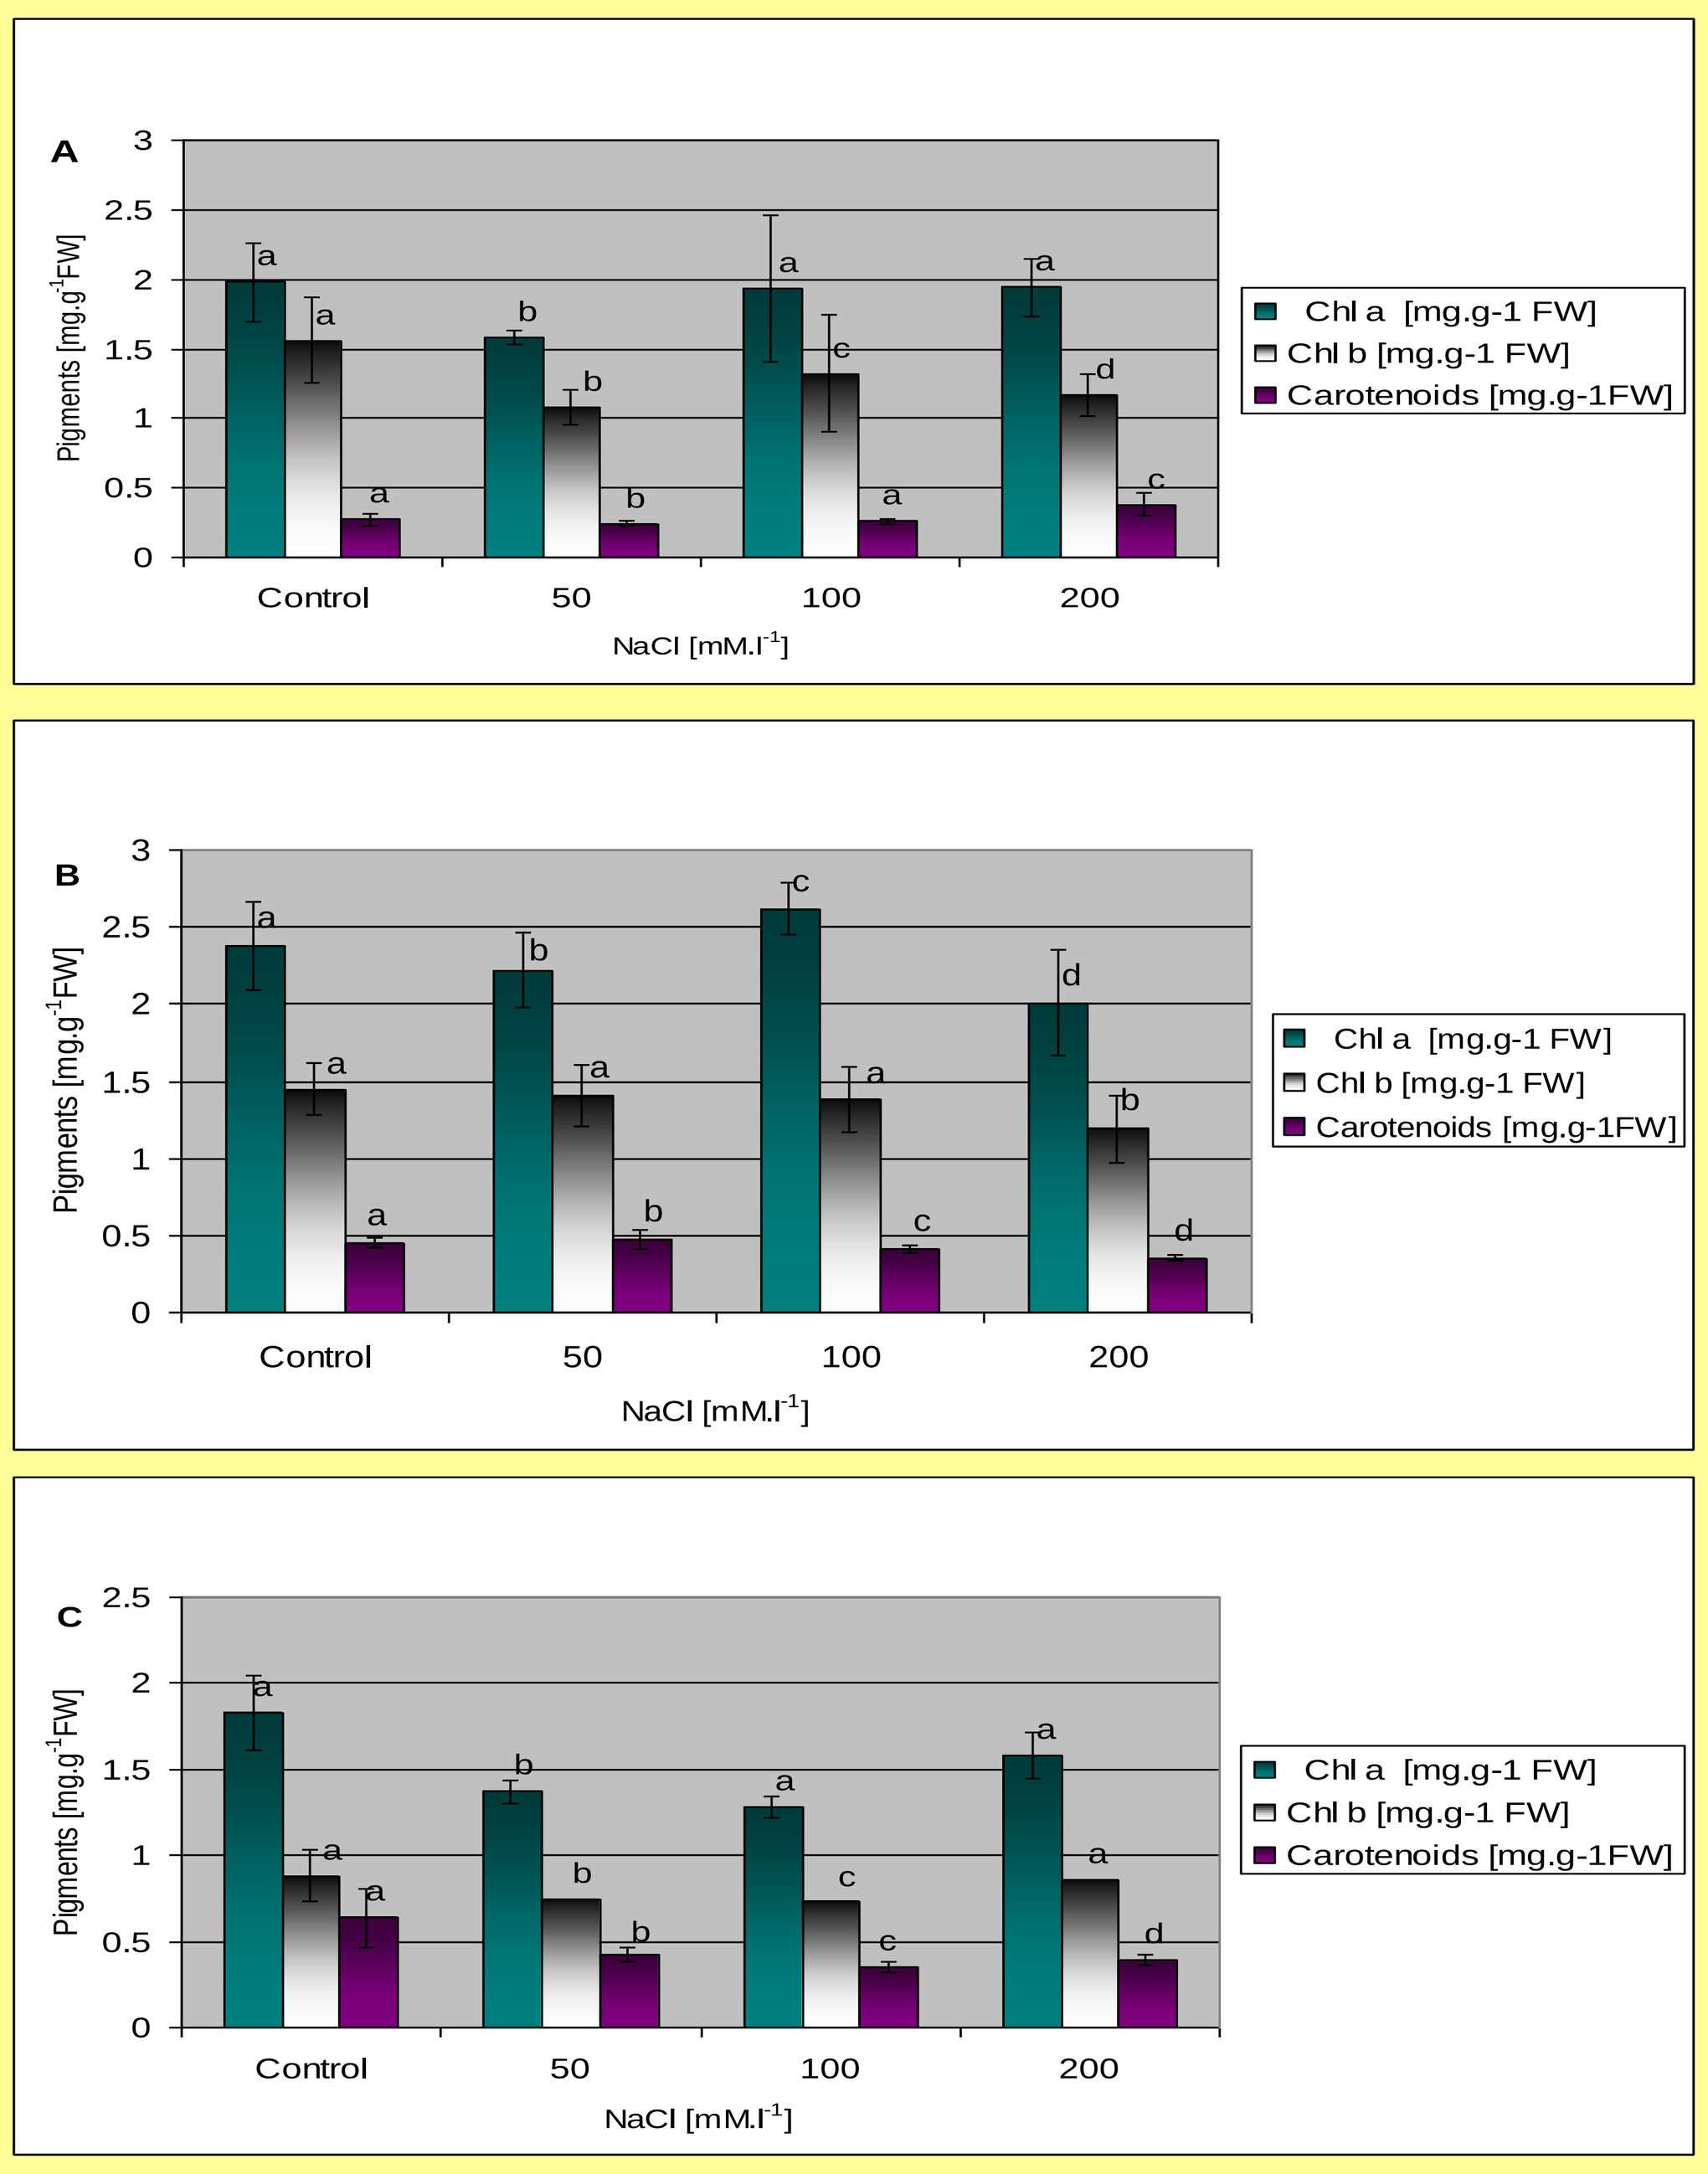

A
B
C
Pigments [mg.g^{-1}FW]
Control
50
100
200
NaCl [mM.l^{-1}]
Chl a [mg.g-1 FW]
Chl b [mg.g-1 FW]
Carotenoids [mg.g-1FW]

Fig. 1 Changes in chlorophyll a, chlorophyll b and carotenoides in the leaves of Paulownia clones clones grouth at 3 levels of salinity.

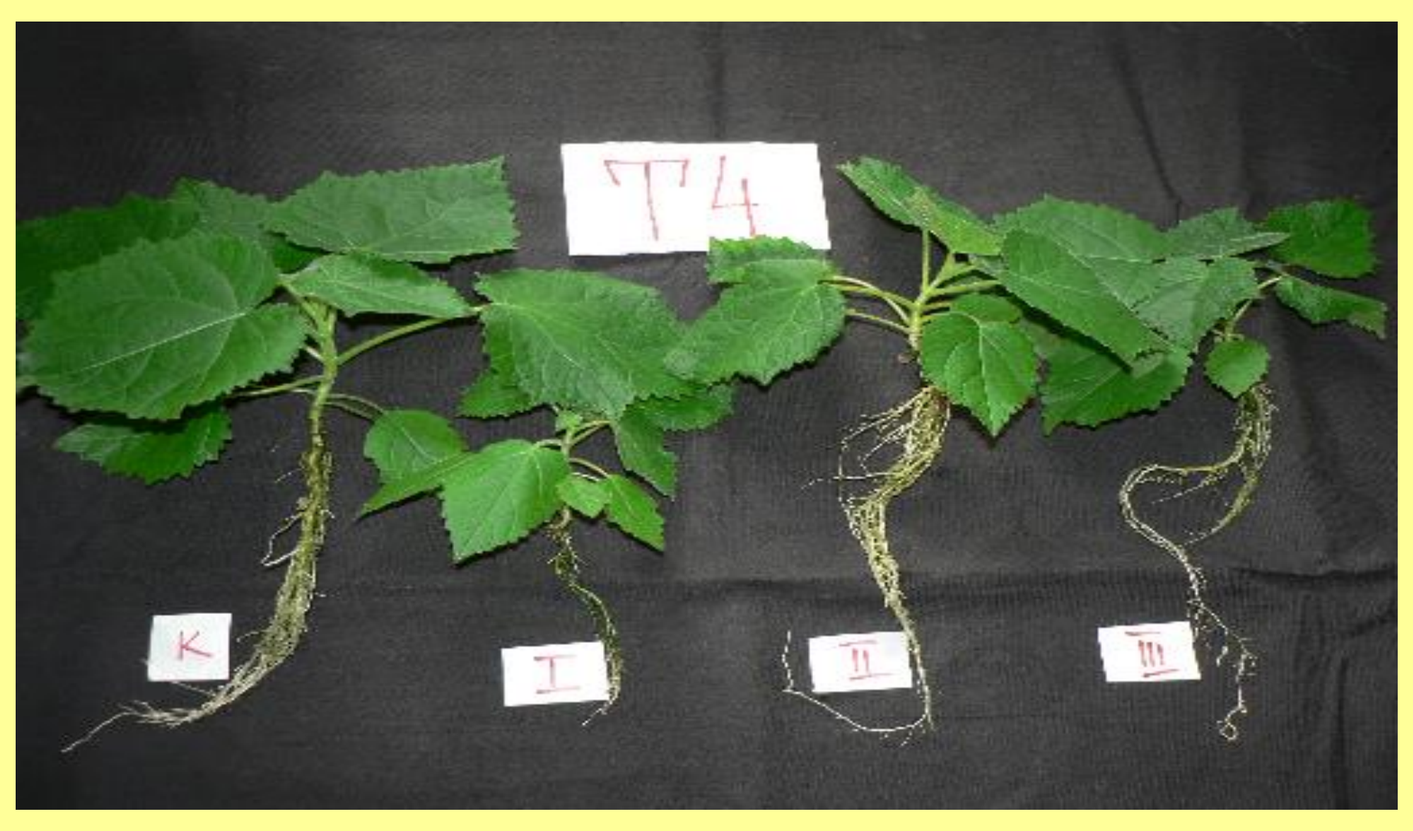

T4
K
I
II
III

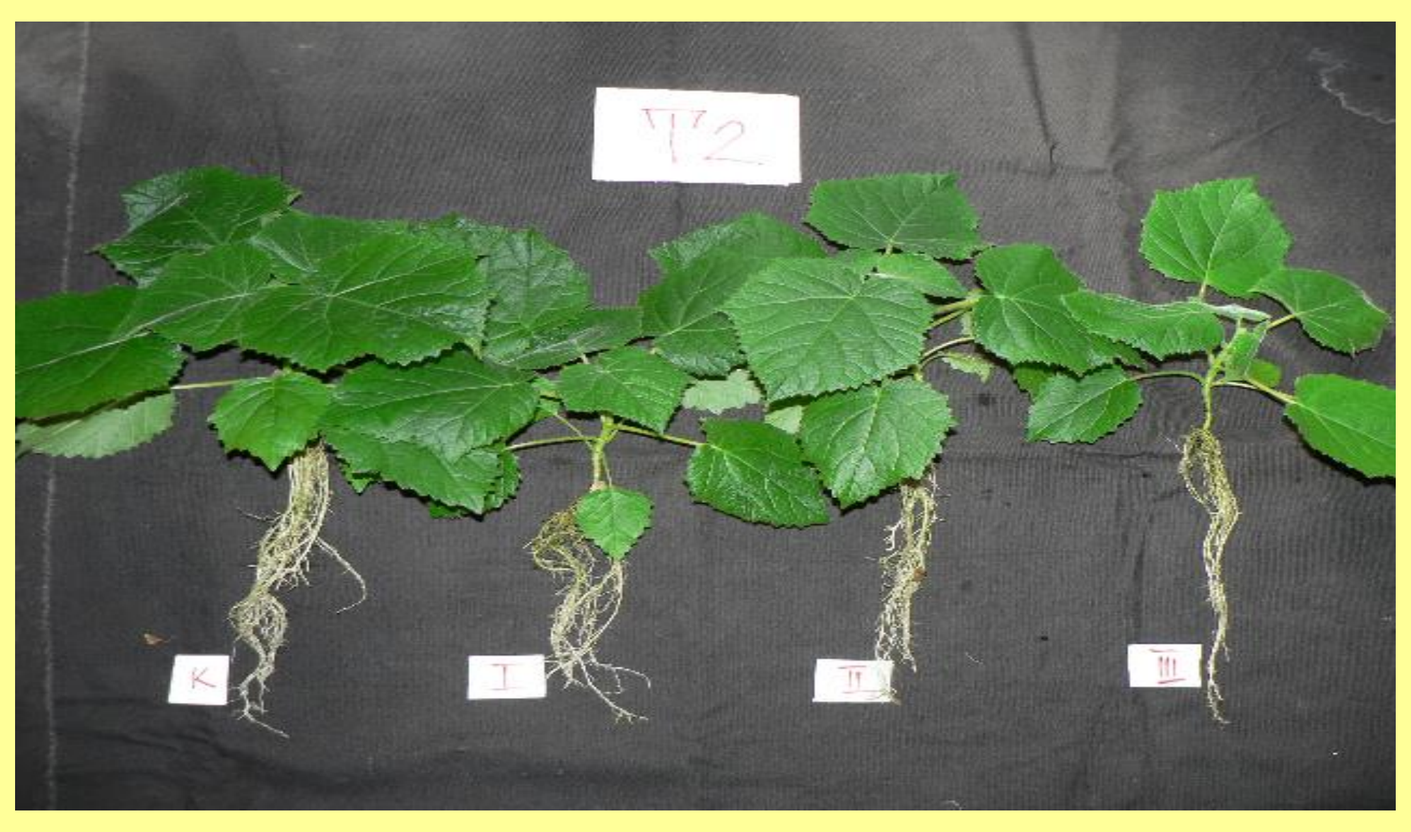

T2
K
I
II
III

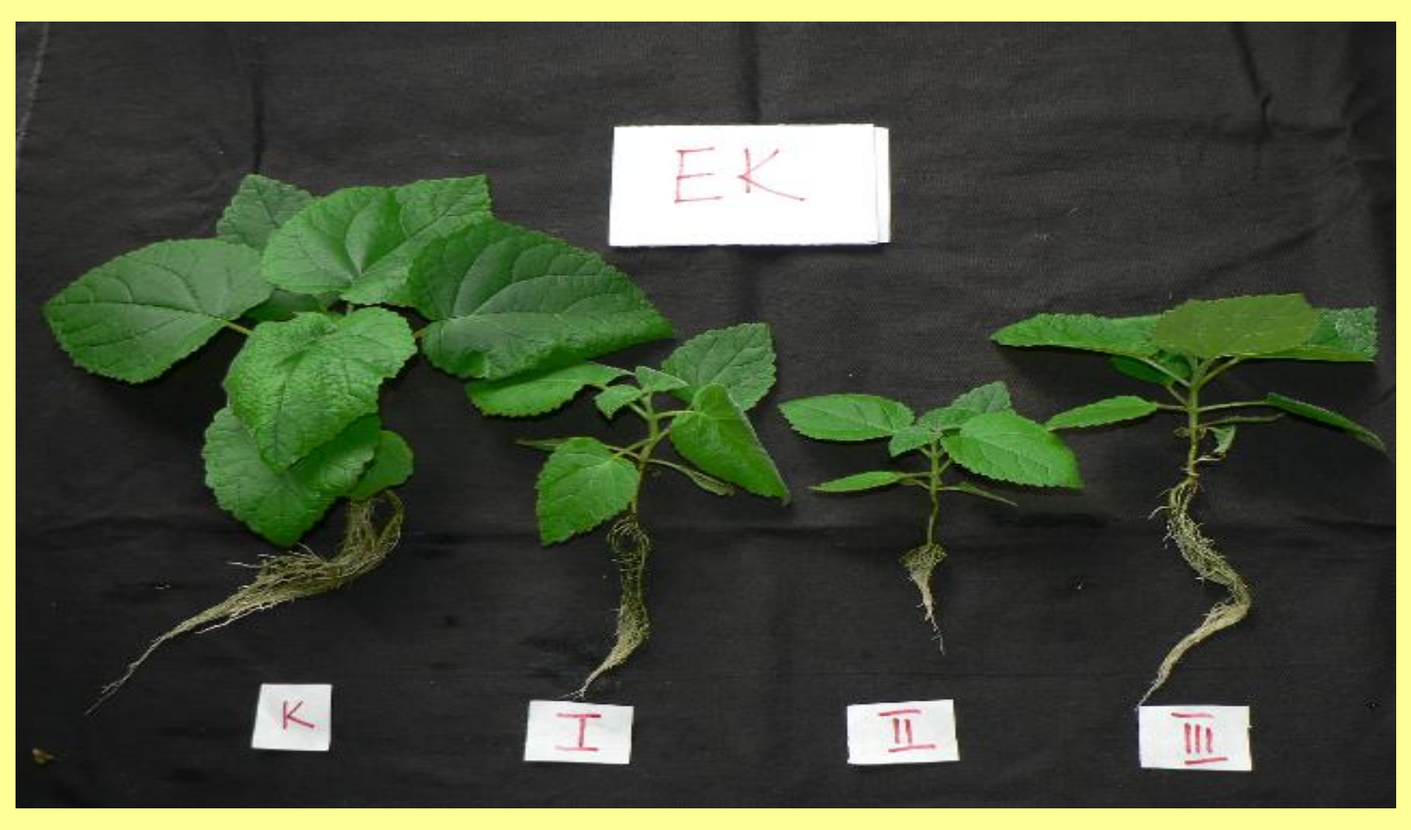

EK
K
I
II
III

RESULTS AND DISCUSSION

Effect of salt stress on plants growth. The seedlings growth is normally limited by increasing concentration of NaCl (Sreenivasulu et al., 2000). In our study, with increasing salinity levels, the root and stem length, leaf number and total leaf area in the three plants were reduced (Table 1). The root and stem length of EK clone was reduced more than that of T4 and T2 clone. The values for stem length measured at 200 mmol.l^{-1} NaCl for T2 clone was higher than control. The leaf number of EK clone rose, but the total leaf area declined sharply with increasing salinity levels. Maximum reduction of the total leaf area was observed at 200 mmol.l^{-1} NaCl by 55% for T4, 42% for EK and 37% for T2, respectively, compared to the control. The total leaf area showed the capability of a plant in forming of photosynthetic surface. The total DW per plant was increased for T4 by 28% and 47% under low salinity levels (50 mmol.l^{-1} and 100 mmol.l^{-1} NaCl), but was decreased by 37% at 200 mmol.l^{-1} NaCl.The total DW per plant were reduced strongly for T2 and EK clones with increasing salinity levels. Under highest salinity level (200 mmol.l^{-1} NaCl) the values decreased about 45% for EK and about 66% for T2 clone, respectively, compared to the control. The roots to shoot dry mass ratios were increased in a different manner with increasing salinity levels. The highest values was observed at 200 mmol.l^{-1} NaCl for T4 clone (~60%), followed by EK clone (~22%) and T2 clone (~3%).

Effect of salt stress on pigment content. Higher salinity reduced the chlorophyll a content in the leaves of three clones. High NaCl concentrations resulted in a significant decrease the chlorophyll b, namely in leaves of T4 and T2 clones (Figure 1). Both pigments decreases were reported in spinach (*Spinacia oleraceae*) exposed to salinity stress (Di Martino et al., 2003). The carotenoid content significantly decreased in the leaves of EK and T2 clones grown in the highest concentrations of NaCl, but increased in the leaves of T4 plants. In sweet cherry rootstock Gisela 5 (*Prunus cerasus x canescens*) a decrease in chlorophyll content was observed as a result of NaCl exposure (0, 50, 100 and 150 mM) (Erturk et al., 2007) and in beans green and yellow pigments decreased with increasing salinity (100 mM NaCl) (Stoeva et al., 2008). Our results showed that the chlorophyll a/chlorophyll b ratios increased in T4 clone whereas green/yellow pigment ratios decreased with increasing salinity levels (Table 2). Reverse tendency was observed in the leaves of EK clone and these parameters changed heterogeneously in the leaves of T2 clone.

During the plant growth and development, the form and functions of organs undergo a considerable change, and the ability of plants to react to salinity stress depends on the genes that are expressed at the stage of development during which the stress is imposed. The mechanism for salinity tolerance becomes even more complicated when response to salinity stress of a plant species varies with the growth stage of its life cycle. In order to develop practicable strategies for selecting salt tolerant clones/genotypes of potential crops, there is a need to gain detailed information on whether changes in physiological and biochemical parameters due to salt stress are attributable to detrimental effects of salt stress, or are components of the adaptation mechanism (Ashraf, 2004; Ashraf et al., 2004).

Our results showed that *Paulownia elongata x fortunei x elongata* - T2 clone is more tolerant to salt stress at the salinity conditions tested than *P. elongata x elongata* - T4 and *P. elongata x kawakarnii* - EK clones. Further investigations are needed to determine the influence of NaCl on *Paulownia* clones, in order to elucidate the mechanisms utilized by these clones. In other studies (Ayala – Astorga et al., 2009; Ayala – Astorga et al., 2010) with *Paulownia imperialis* and *Paulownia fortunei* species grown *in vitro*, it is reported they survived at 60 mM NaCl concentration added to culture medium, and higher concentrations induced necrosis and death. Obtained results are not compared because *Paulownia* clones are imposed to increasing salinity levels in hydroponic experiment whereas the other plants are *in vitro* grown at different NaCl concentrations.

In general there is much work to do to improve crops because the problem is increasing with salinity, drought, and other worldwide factors that affect crops. We are considering expanding our research on *Paulownia* clones, produced by BioTree Ltd., Bulgaria since it is important woody species in reforestation programs, which can diminish desertification in many regions of Bulgaria and will be an intercropping species.

Introduction

The environmental degradation, mainly soil pollution and erosion cause biotic and abiotic stress in plants and limited their growth and development (Shao et al., 2005). Among abiotic stresses drought and salinity are major stresses that harm crop losses worldwide (Vinocur et al., 2005). High salinity causes severe damage to plants, including growth inhibition, impaired metabolism, necrosis, and loss of production and quality (Sivritepe et al., 1999). The research to improve salt tolerance in plants is mainly focused on biochemical and physiological aspects and the genes responsible for salt tolerance in some species have been identified (Schleiff, 2008). Stress might occur as a complex mechanism of several interacting environmental factors that cause variations in plant phenotypes, as plants respond to complex growth conditions (Shao et al., 2007). The plant cell and tissue culture methods could be useful in studying the salinity tolerance mechanisms in plants and their effects on crop production when are not evidently known (Akinci et al., 2004).

Paulownia is native from China. Paulownia tomentosa has been introduced into USA and Europe as an ornamental plant and is still widely used for this purpose. Trees introduced in Bulgaria reach 12 m average height and 13.4 cm average diameter during 7 years (Kalmukov, 1995). Paulownia Sieb.&Zucc. are used for afforestation in Eastern Black Sea Region of Turkey due to high adaptability, fast growth and heavy leaf biomass (Yavuzsefik et al., 2001). Over the last two decades Paulownia species has been extensively studied due to its ability to uptake nitrates and land contaminants, namely heavy metals (Doumett et al., 2008; Azzarello et al., 2012). This high-yielding tree can be used for the production of energy, paper pulp and wooden building materials. The genetically tissue–cultured Paulownia seedlings produced by The World Paulownia Institute (WPI) allow production of biofuels after introducing of cultivars without detrimental impacts on food supply or the environment. Research on in vitro propagation of P. elongata and P. fortunei has been reported (Bergmann et al., 1997). Application of this technology for micropropagation of tree species offers a rapid means of producing clonal planting stock for afforestation, woody biomass production and it is effective way to maintain the genetic gain (Park et al., 1992).

Plants used in the current paper are propagated and rooted according technology registered by BioTree Ltd., Bulgaria. This laboratory is largest producer and supplier of genetically superior Paulownia tissue -cultures – in vitro seedlings.

In this research, the effect of NaCl on the growth and plastid pigments content in leaves of three Paulownia clones (P. elongata x fortunei x elongata - T2, P. elongata x elongata - T4, P. elongata x kawakarnii - EK), grown as hydroponic culture after transplantation the explants were compared so as to provide fundamental base for vegetation restoration in salinized soils.